Abstract 186

Thromboembolic Events in Hospitalized Cancer Patients: Impact on Stay Duration and Cost for Four Major Cancer Localizations

Florian SCOTTE[a], Cécile BLEIN[b], Nicolas MARTELLI[c], Isabelle BORGET[d]

a. Medical Oncology and Supportive Care Unit, Georges Pompidou Hospital, Paris; Ethics Resarch Unit EA 4569, Paris Descartes University, Paris. - b. Heva, Lyon
c. Pharmacy, Georges Pompidou Hospital, Paris - d. Research and Study in Health Economics, Institut Gustave Roussy, Villejuif; Pharmaceutical Law and Economics Chair, Paris-Sud University, Chatenay-Malabry

ABSTRACT

Background:
Cancer patients represent an at-risk population for Venous Thromboembolic Events (VTE). Our study aimed to evaluate the impact of VTE on the length and cost of hospital stay in French patients hospitalized for breast cancer (BC), colon cancer (CC), lung cancer (LC) or prostate cancer (PC).

Methods:
The French national hospital database (PMSI) and the disease-specific ICD-10 codes were used to identify BC, CC, LC or PC patients diagnosed in 2010 who were hospitalized with a VTE during the following two years. We selected stays in public hospitals during which a VTE occurred but was not the main reason of hospitalization (cancer was classified as primary/related diagnosis and VTE as significant associated diagnosis). Those stays were matched and compared to similar stays (same cancer and same reason for hospitalization) without VTE. Costs were calculated using the French official tariffs, from the perspective of the third-party payer.

Results:
We identified 214 stays for breast cancer during which a VTE occurred and was classified as significant associated diagnosis, 843 stays for colon cancer, 1,301 for lung cancer and 126 for prostate cancer.
The comparison between those stays and similar stays without VTE showed a significant increase of hospital stay duration in patients experiencing VTE. Median duration raised from 4 to 7 days in BC patients, from 8 to 16 days in CC, from 2 to 9 days in LC and from 6 to 10 days in PC.
Consequently, the median expenditure per stay increased by 37% in BC patients with VTE (up to € 5,518), by 61% in CC (up to € 9,878), by 202% in LC (up to € 7,308) and by 22% in PC (up to € 6,200).

Conclusion:
When occurring during hospitalization, VTE made cancer management much heavier: patients faced prolonged hospital stays whereas healthcare system faced significant additional cost. Better prevention and follow-up measures could reduce this burden, and benefit both patients and hospitals.

INTRODUCTION

- In France, cancer-related costs represent a growing part, namely 10% of third-party payer expenses in 2012 (€ 14.5 billion) with an increase of 2.8% compared to 2011. All available options to reduce or at least stabilize the costs without decreasing the quality of patient care must be considered[1].
- Patients with cancer are known to have a four-to-six fold increased risk of venous thromboembolism (VTE), including deep venous thrombosis and pulmonary embolism[2,3]. These venous thromboembolic events lead to hospitalizations that dramatically increase the burden of patients' management.
- Several international studies tried to estimate the VTE burden in cancer patients: in the USA, an observational study of patients treated with chemotherapy showed a VTE overall incidence of 13.5% at 12 months[4]. In addition, the risk of recurrence in cancer patients has been estimated at 20.6%, compared to 6.8% in patients without cancer[5]. A study in ambulatory US patients with various types of cancer revealed that patients with VTE had a total healthcare cost increased by 79.9%, compared to those without VTE over the 12-month post VTE follow-up period[6].
- Our previous study regarding two types of cancer (breast and prostate) demonstrated that VTEs have significant costs when leading to hospitalizations as primary or related diagnosis[7]. In our knowledge, there is a lack of data concerning VTE occuring as a co-morbidity during cancer related hospitalizations.

AIMS OF THE STUDY

- To extend the previous results obtained in breast and prostate cancers, to colon and lung cancers.
- To evaluate the epidemiological and economical burden of cancer-related VTE in France in four of the most frequent types of cancer:
 - To estimate the number of hospital admissions related to VTE in newly diagnosed breast, colon, lung or prostate cancers patients;
 - To determine VTE-recurrence impact;
 - To assess the impact of VTE on stay duration and costs, when VTE occurred as SAD in patients hospitalized for cancer.

METHODS

- **Data source:**
 The French Medical Information System (Programme de Médicalisation des Systèmes d'Information: PMSI), which covers all French public and private hospitals. For each patient, PMSI contains: diagnosis that led to hospitalization, associated diagnoses related to co-morbidities and/or complications during admission and examinations performed during the admission.
- **Data collection of VTE-related admissions:**
 Cases were extracted from the PMSI database using the International Classification of Diseases, 10th revision (ICD-10 codes). We kept patients with breast cancer (code C50), colon cancer (code C18), lung cancer (code C34) or prostate cancer (code C61) and selected newly diagnosed patients in 2010 (incident cancer cases).
 Among these new BC, CC, LC or PC patients, we searched for those hospitalized with DVT (code I80) or PE (code I26) as primary diagnosis (PD), related diagnosis (RD) or significant associated diagnosis (SAD). Incident patients with DVT or PE were followed for 2 years (2011 and 2012) using PMSI database, in order to identify the number of thromboembolic recurrences that required hospitalization.
- **Number of stays and patients hospitalized for VTE:**
 The number of hospital stays when a VTE event occurred during the 2 years follow-up period was estimated, as well as the corresponding number of patients hospitalized at least once.
- **Economic evaluation:**
 Hospital costs were estimated from a healthcare payer's perspective. Costs of chemotherapy, ambulatory and indirect costs related to productivity loss were not considered in the analysis. Expenses were calculated using the official 2010, 2011 and 2012 public and private hospitals Diagnosis Related Groups (DRG) tariffs, which included nursing care, treatments, drugs, accommodation and investment costs, as well as medical and technical acts.
- **Comparison between hospital stays with and without VTE occurrence:**
 In the second part of our study, we numbered hospital stays for cancer as PD/RD and distinguished those during which a VTE occurred and was classified as SAD from those without VTE. Differences in distribution of age, gender and hospitalization type (public or private) were observed between the two cohorts. To reduce the impact of those factors, the data from both groups were paired: the cohort without VTE was randomly reconstituted according to the age, gender and hospitalization type of the patients with VTE.
 Duration and cost of these stays as SAD were then compared to similar stays without VTE occurring. Median and mean durations of hospital stays as well as median costs were estimated. Statistical significativity tests were achieved on median values.

RESULTS

1 Incidence of VTE-related hospitalization

Among all breast, colon, lung and prostate cancer patients hospitalized in 2010, we took out patients who had been hospitalized for those cancers from 2006 to 2010. Newly diagnosed cases in 2010 represented a population of 194,964 patients. 62,365 patients were newly diagnosed with breast cancer, 47,594 with colon cancer, 39,454 with lung cancer and 45,551 for prostate cancer.
During a 2-year follow-up period, 8,909 patients were hospitalized for a VTE (or presented a VTE during an hospitalization): 1,271 breast cancer patients (2.04% of incident cases), 2,866 colon cancer patients (6.02%), 3,775 lung cancer patients (9.57%) and 997 prostate cancer patients (2.19%) (Figure 1). These results are consistent with literature[8,9].

Figure 1. Incidence of VTE-related hospitalizations (%) in newly diagnosed cases

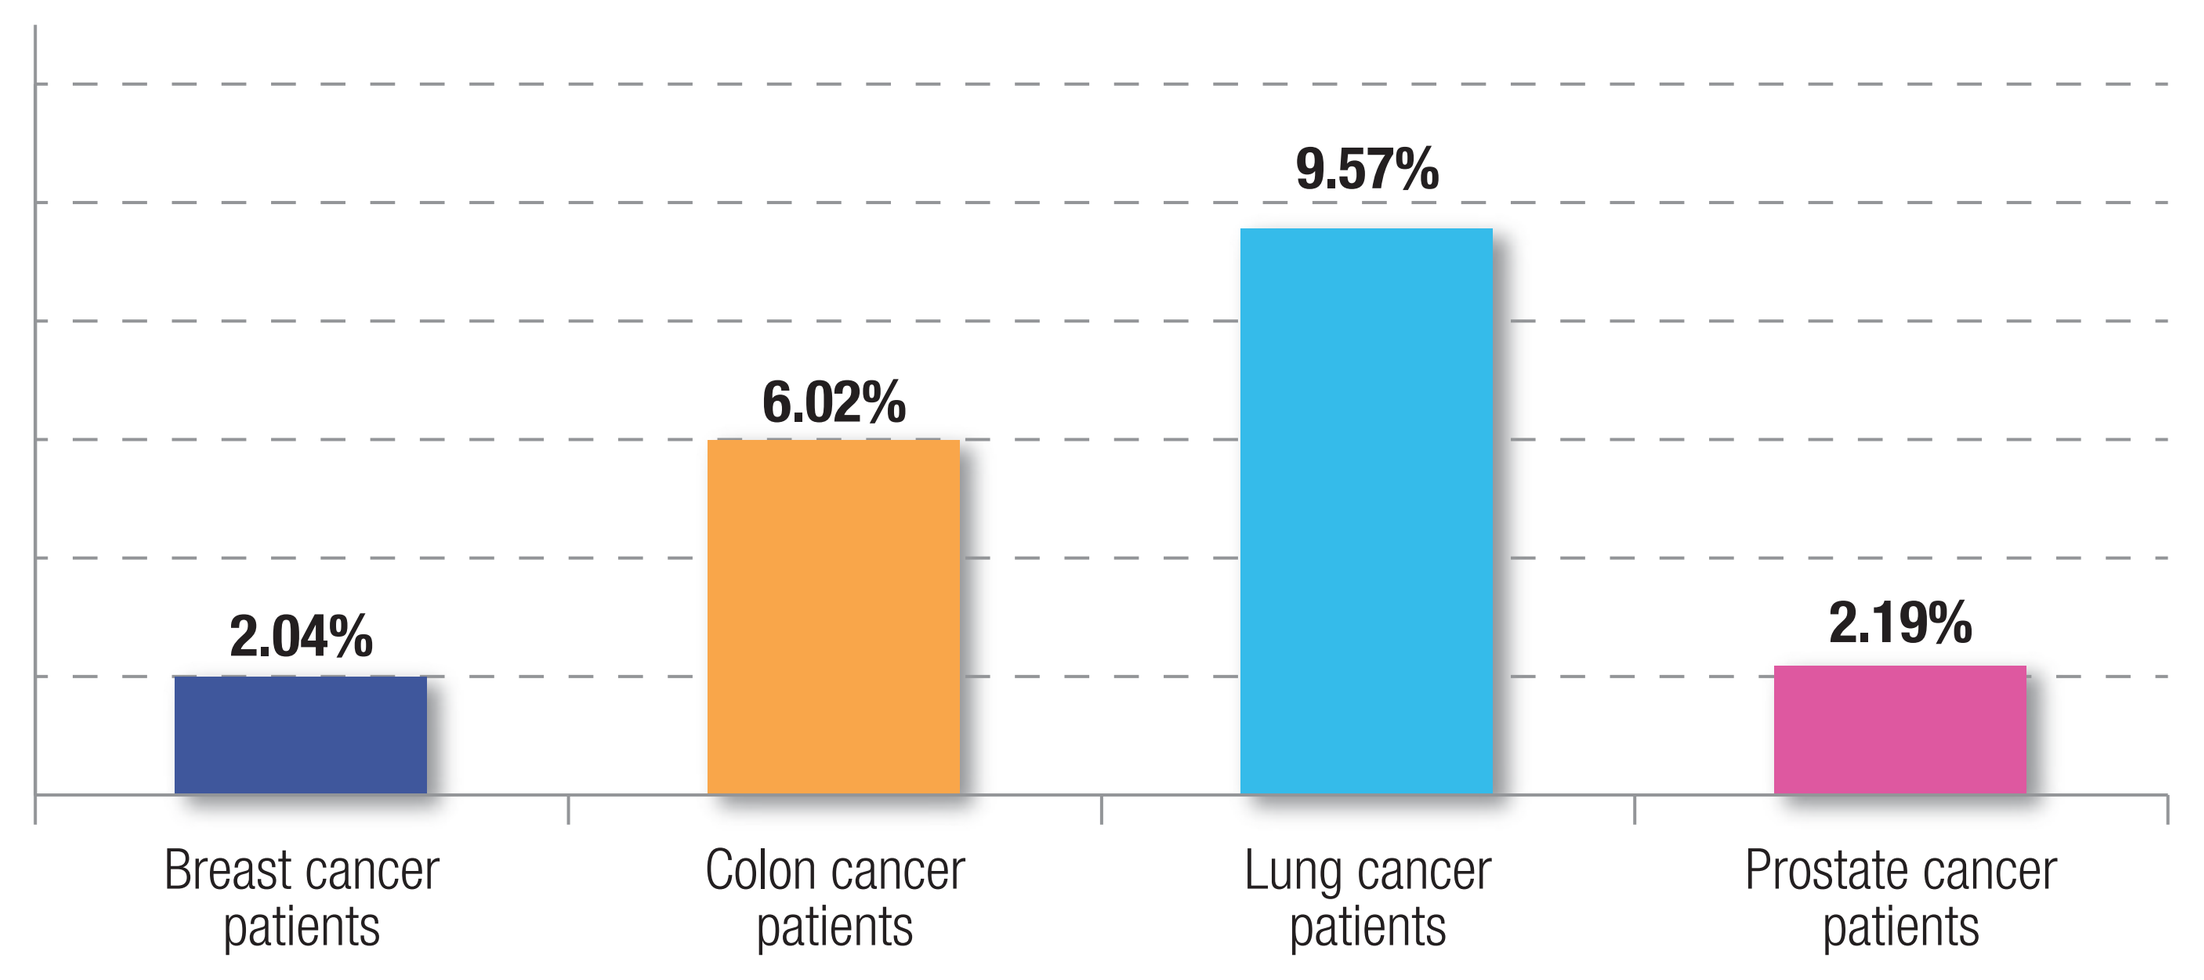
2.04%
6.02%
9.57%
2.19%
Breast cancer patients
Colon cancer patients
Lung cancer patients
Prostate cancer patients

2 VTE recurence

Among all 8,909 patients with VTE, 2,053 (23.0%) were hospitalized for a recurrent event (Figure 2), including 202 (15.9%) breast cancer patients, 639 (22.3%) colon cancer patients, 1,068 (28.3%) lung cancer patients and 144 (14.4%) prostate cancer patients (table 1).

Table 1. Number of patients with recurrent VTE
(VTE as primary, related and significant associated diagnosis)

	Patients with only one VTE	Patients with recurrent VTE	Percentage of patients with recurrent VTE
Breast cancer	1,069	202	**15.9%**
Colon cancer	2,227	639	**22.3%**
Lung cancer	2,707	1,068	**28.3%**
Prostate cancer	853	144	**14.4%**

Figure 2. Total number of patients with recurrent VTE
(VTE as primary, related and significant associated diagnosis)

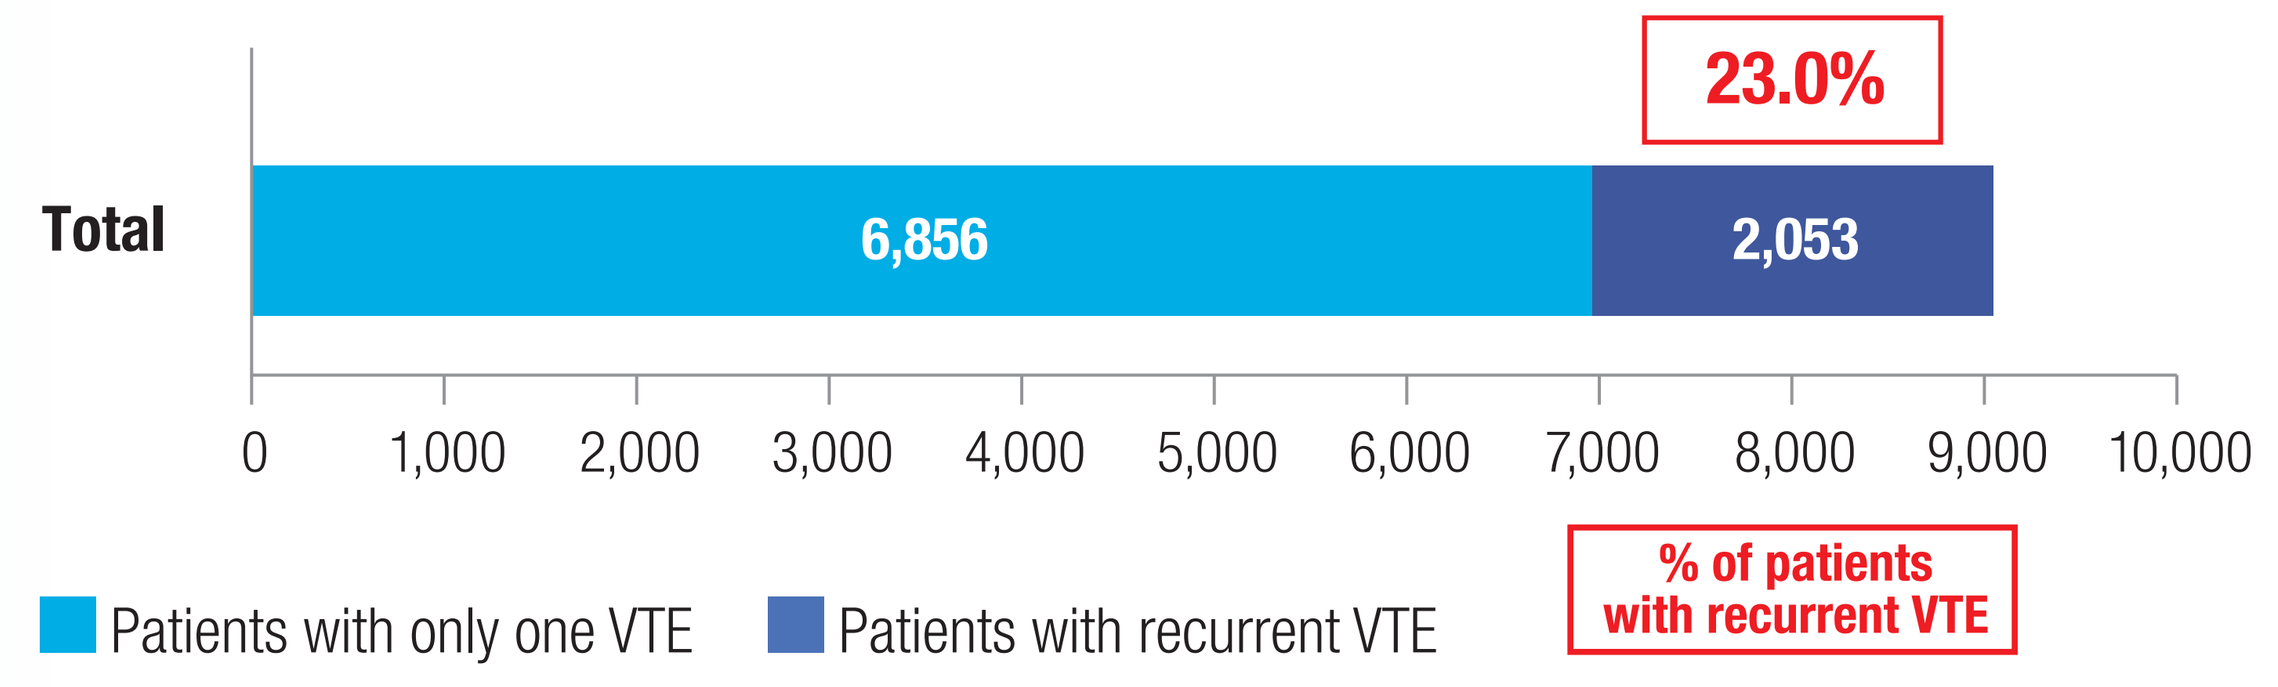
23.0%
Total
6,856
2,053
0
1,000
2,000
3,000
4,000
5,000
6,000
7,000
8,000
9,000
10,000
Patients with only one VTE
Patients with recurrent VTE
% of patients with recurrent VTE

For the patients with VTE, we identified 12,880 hospital stays. 3,969 admissions (30,8%) were therefore due to a recurrent VTE (Figure 3). The mean number of stays per patient with VTE was 1.45.

Figure 3. Total of admissions for recurrent VTE
(VTE as primary, related and significant associated diagnosis)

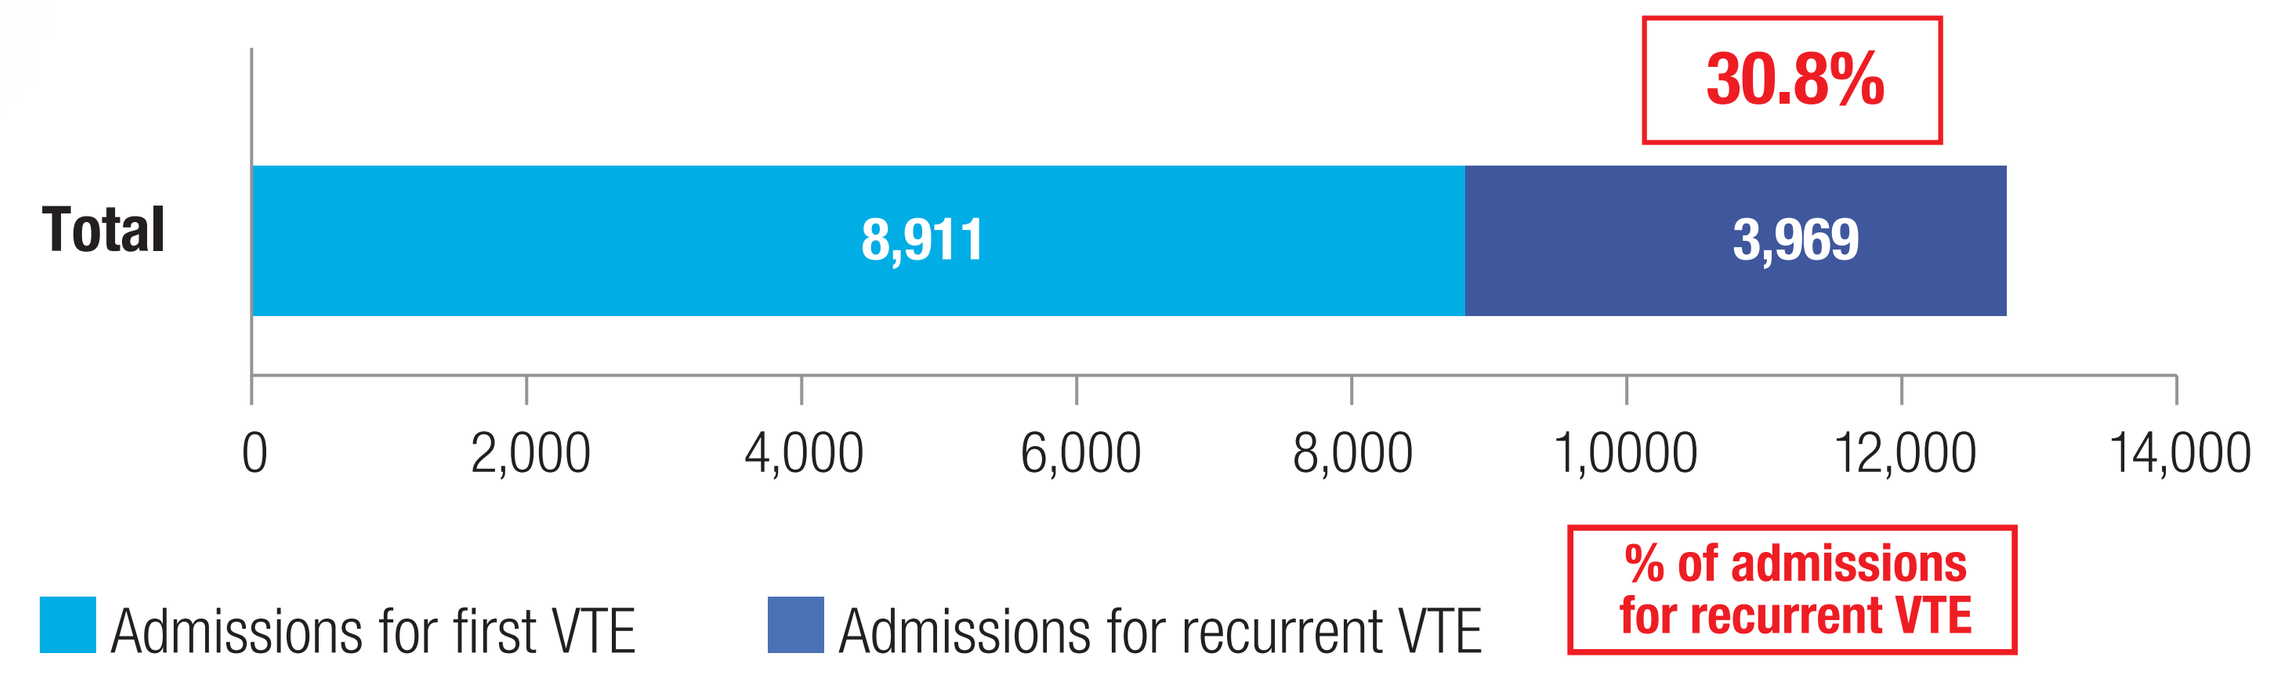
30.8%
Total
8,911
3,969
0
2,000
4,000
6,000
8,000
1,0000
12,000
14,000
Admissions for first VTE
Admissions for recurrent VTE
% of admissions for recurrent VTE

Median duration from the first event to recurrent VTE has been identified for the four types of cancer (Figure 4). It ranged from 19 to 25 days depending on the cancer localization.

Figure 4. Median duration from the first event to rehospitalization for recurrent VTE (days)

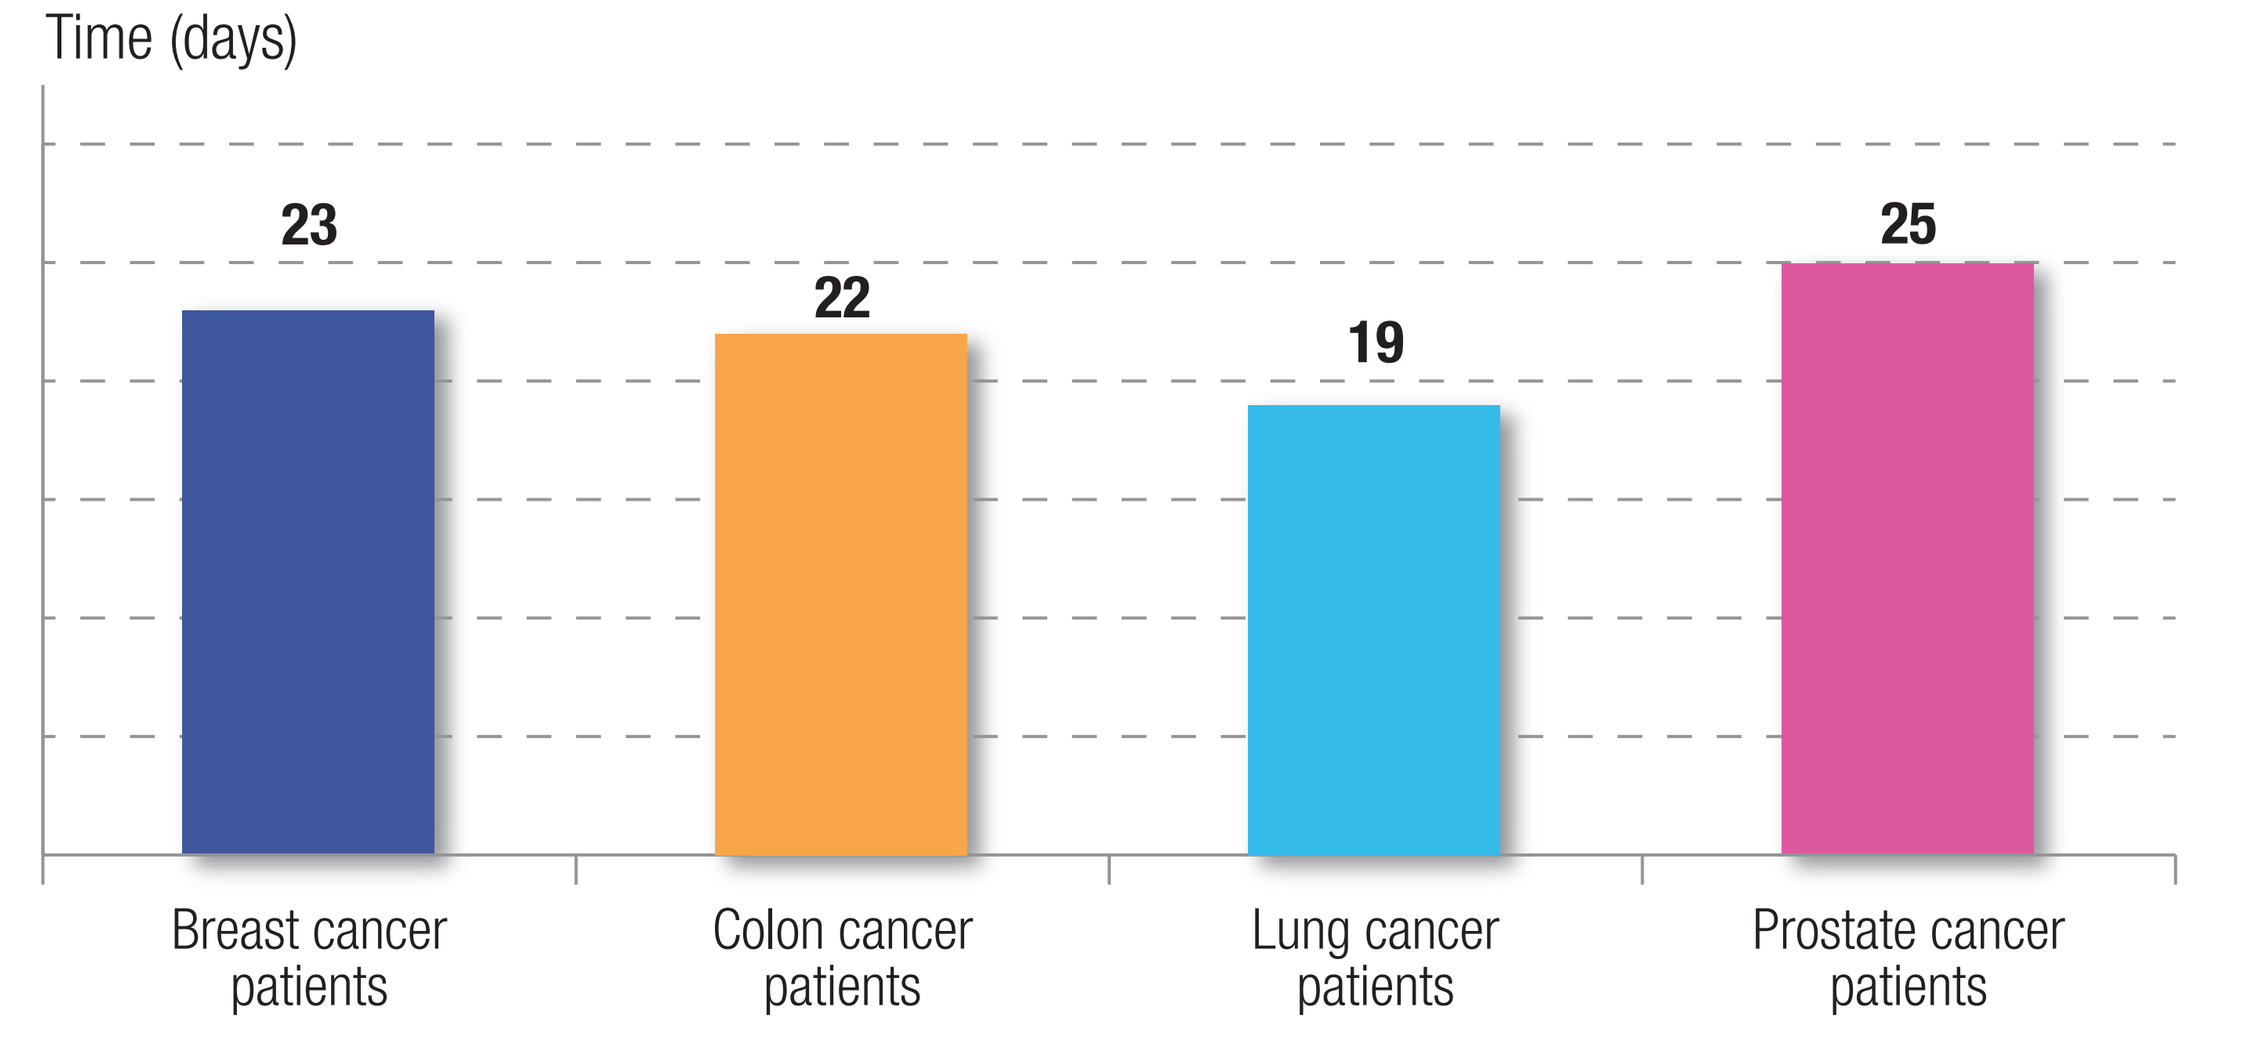
Time (days)
23
22
19
25
Breast cancer patients
Colon cancer patients
Lung cancer patients
Prostate cancer patients

3 VTE occurrence during hospitalization for cancer: impact on stay duration and costs.

We identified 214 stays for breast cancer during which a VTE occurred and was classified as significant associated diagnosis, 843 stays for colon cancer, 1,301 for lung cancer and 126 for prostate cancer. Cancer was classified as PD/RD. In these study, VTE occurrence dramatically prolonged the duration of hospitalization compared to paired cancer patients without VTE, regarding the four cancers. Median stay duration was significatively raised from 4 to 7 days in breast cancer, from 8 to 16 days in colon cancer, from 2 to 9 days in lung cancer and from 6 to 10 days in prostate cancer. (Figure 5).

Figure 5. Median stay duration with and without VTE occurrence (days)
(in public hospitals, cancer classified as PD/RD, VTE as SAD)

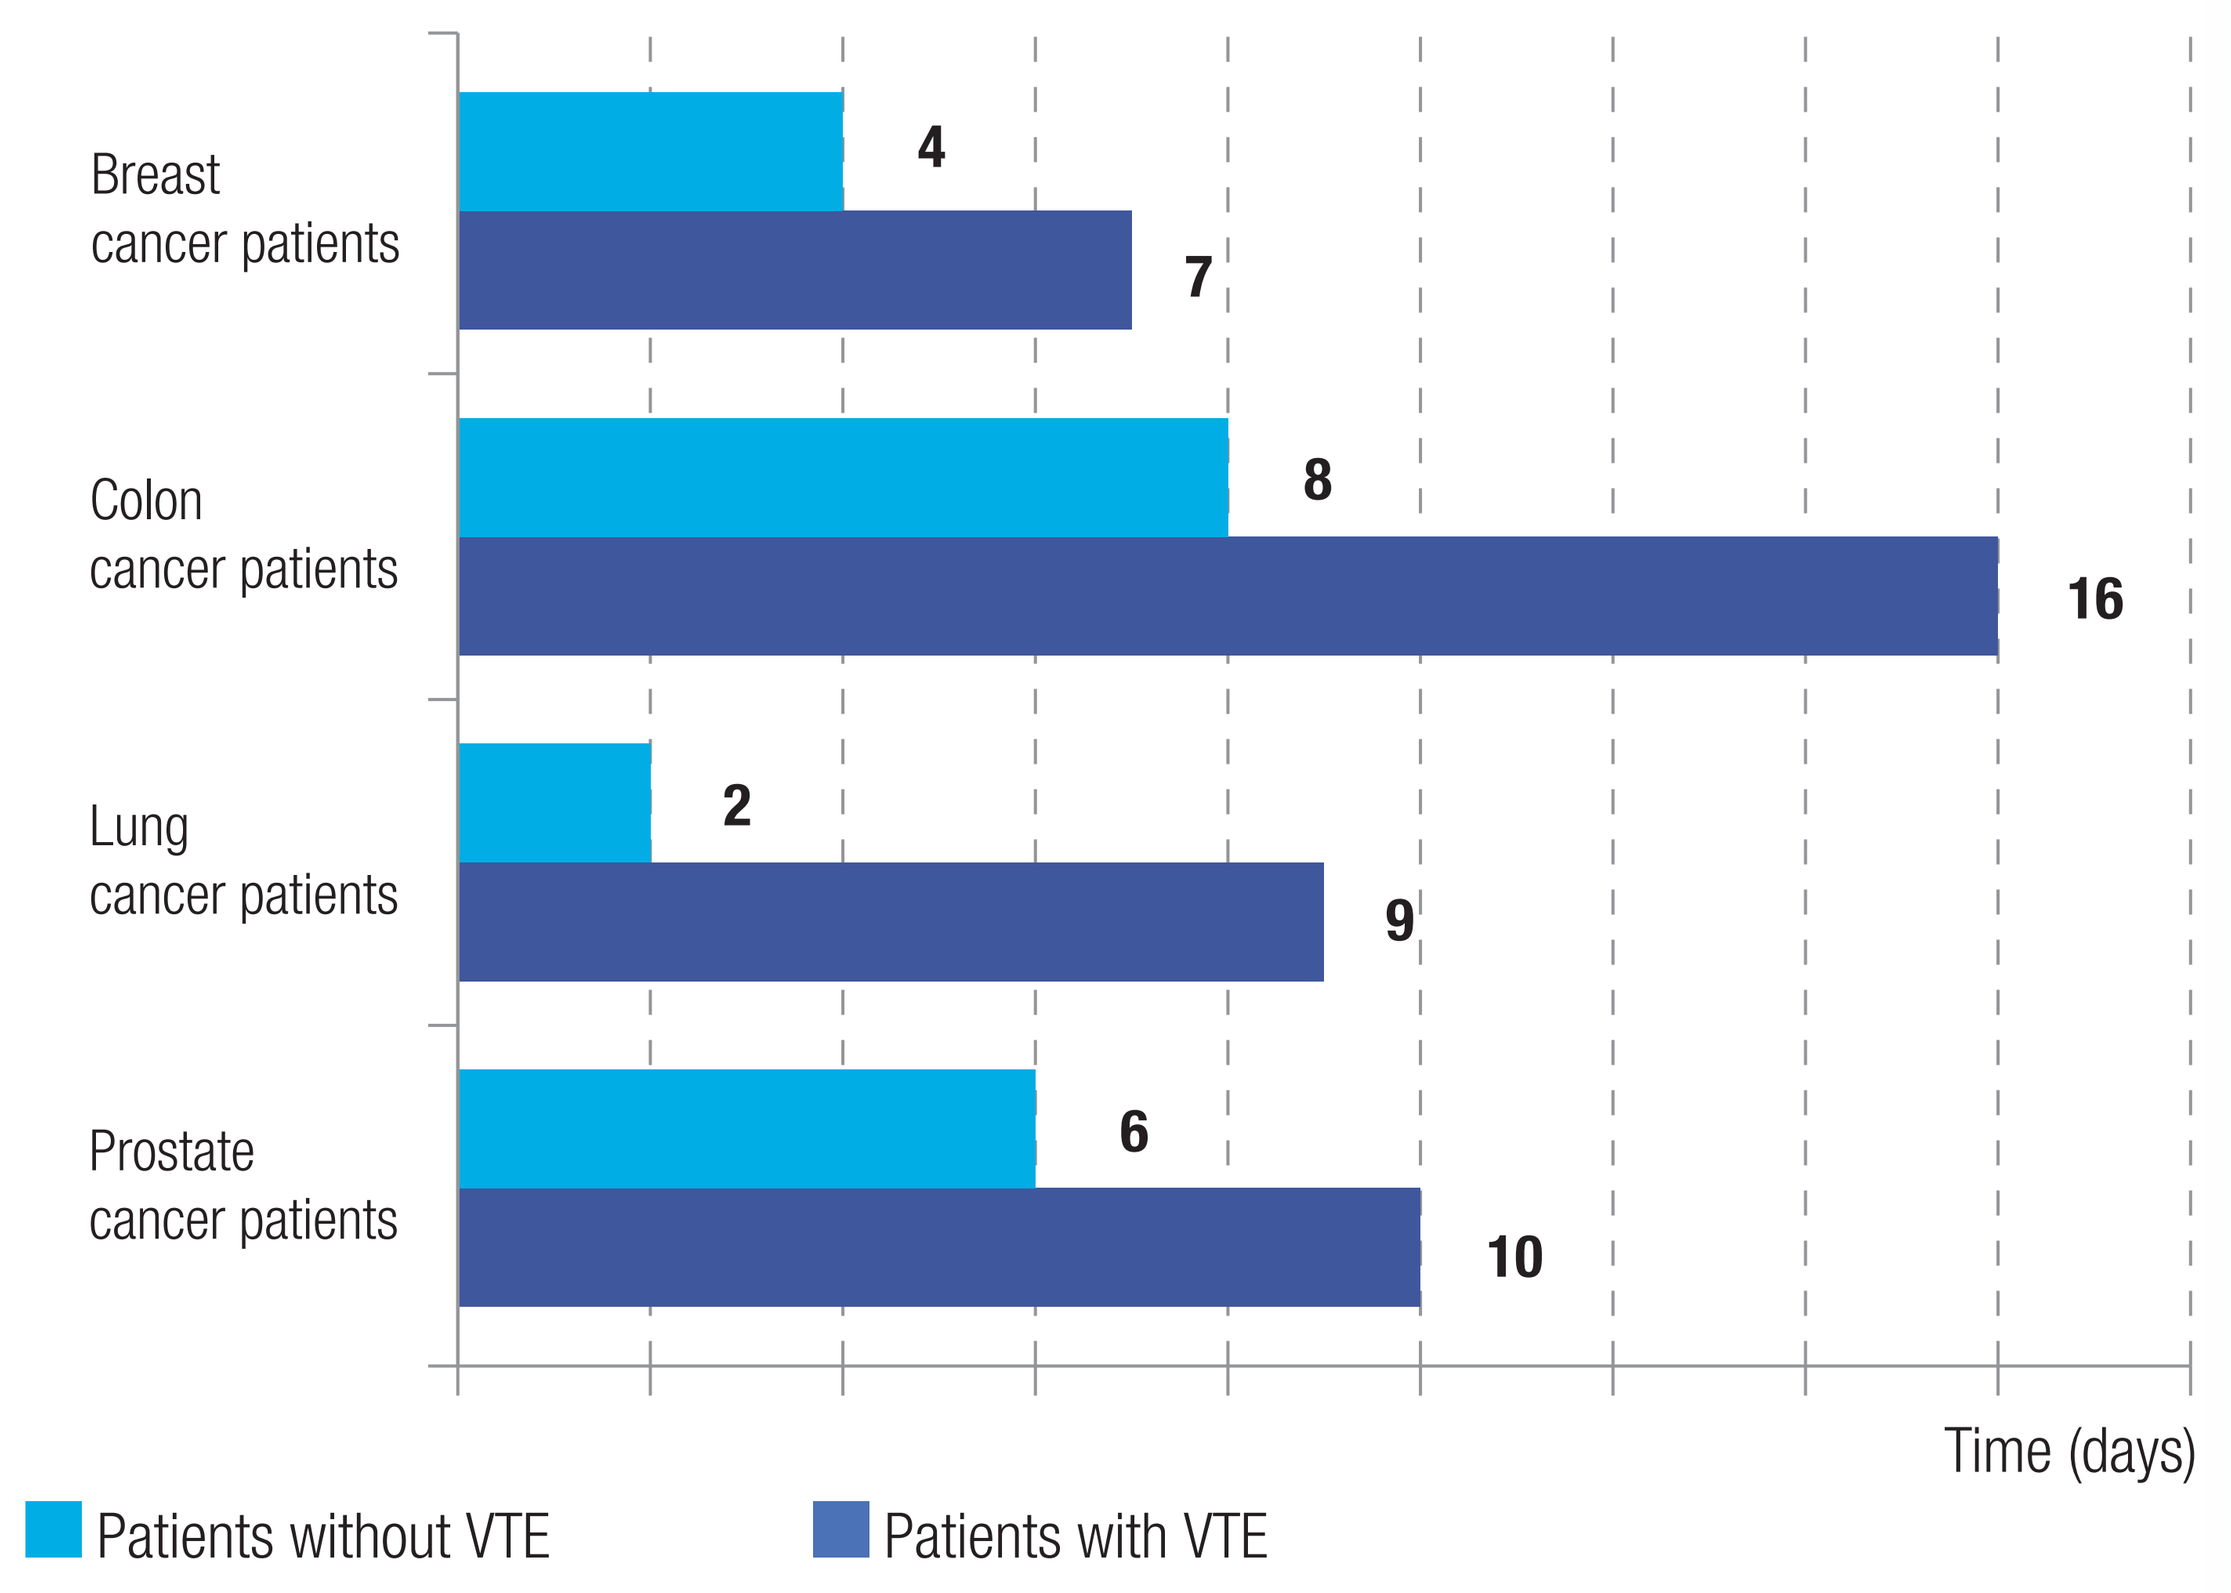
Breast cancer patients
4
7
Colon cancer patients
8
16
Lung cancer patients
2
9
Prostate cancer patients
6
10
Time (days)
Patients without VTE
Patients with VTE

As expected, hospitalization cost was higher for stays during which a VTE occurred. The median amount per stay was significatively increased from € 4,018 to € 5,518 in breast cancer patients, from € 6,171 to € 9,878 in colon cancer patients, from € 2,422 to € 7,308 in lung cancer patients and from € 5,094 to € 6,200 in prostate cancer patients. Therefore, VTE occurrence induced an important extra hospital-cost compared to similar stays without VTE occurrence (patients hospitalized on the same period of analysis). In other words, the median expenditure per stay increased by 37% in breast cancer, by 61% in colon cancer, by 202% in lung cancer and by 22% in prostate cancer (Figure 5).

Figure 5. Median cost per stay for cancer with and without VTE occurrence
(in public hospitals, cancer classified as PD/RD, VTE as SAD)

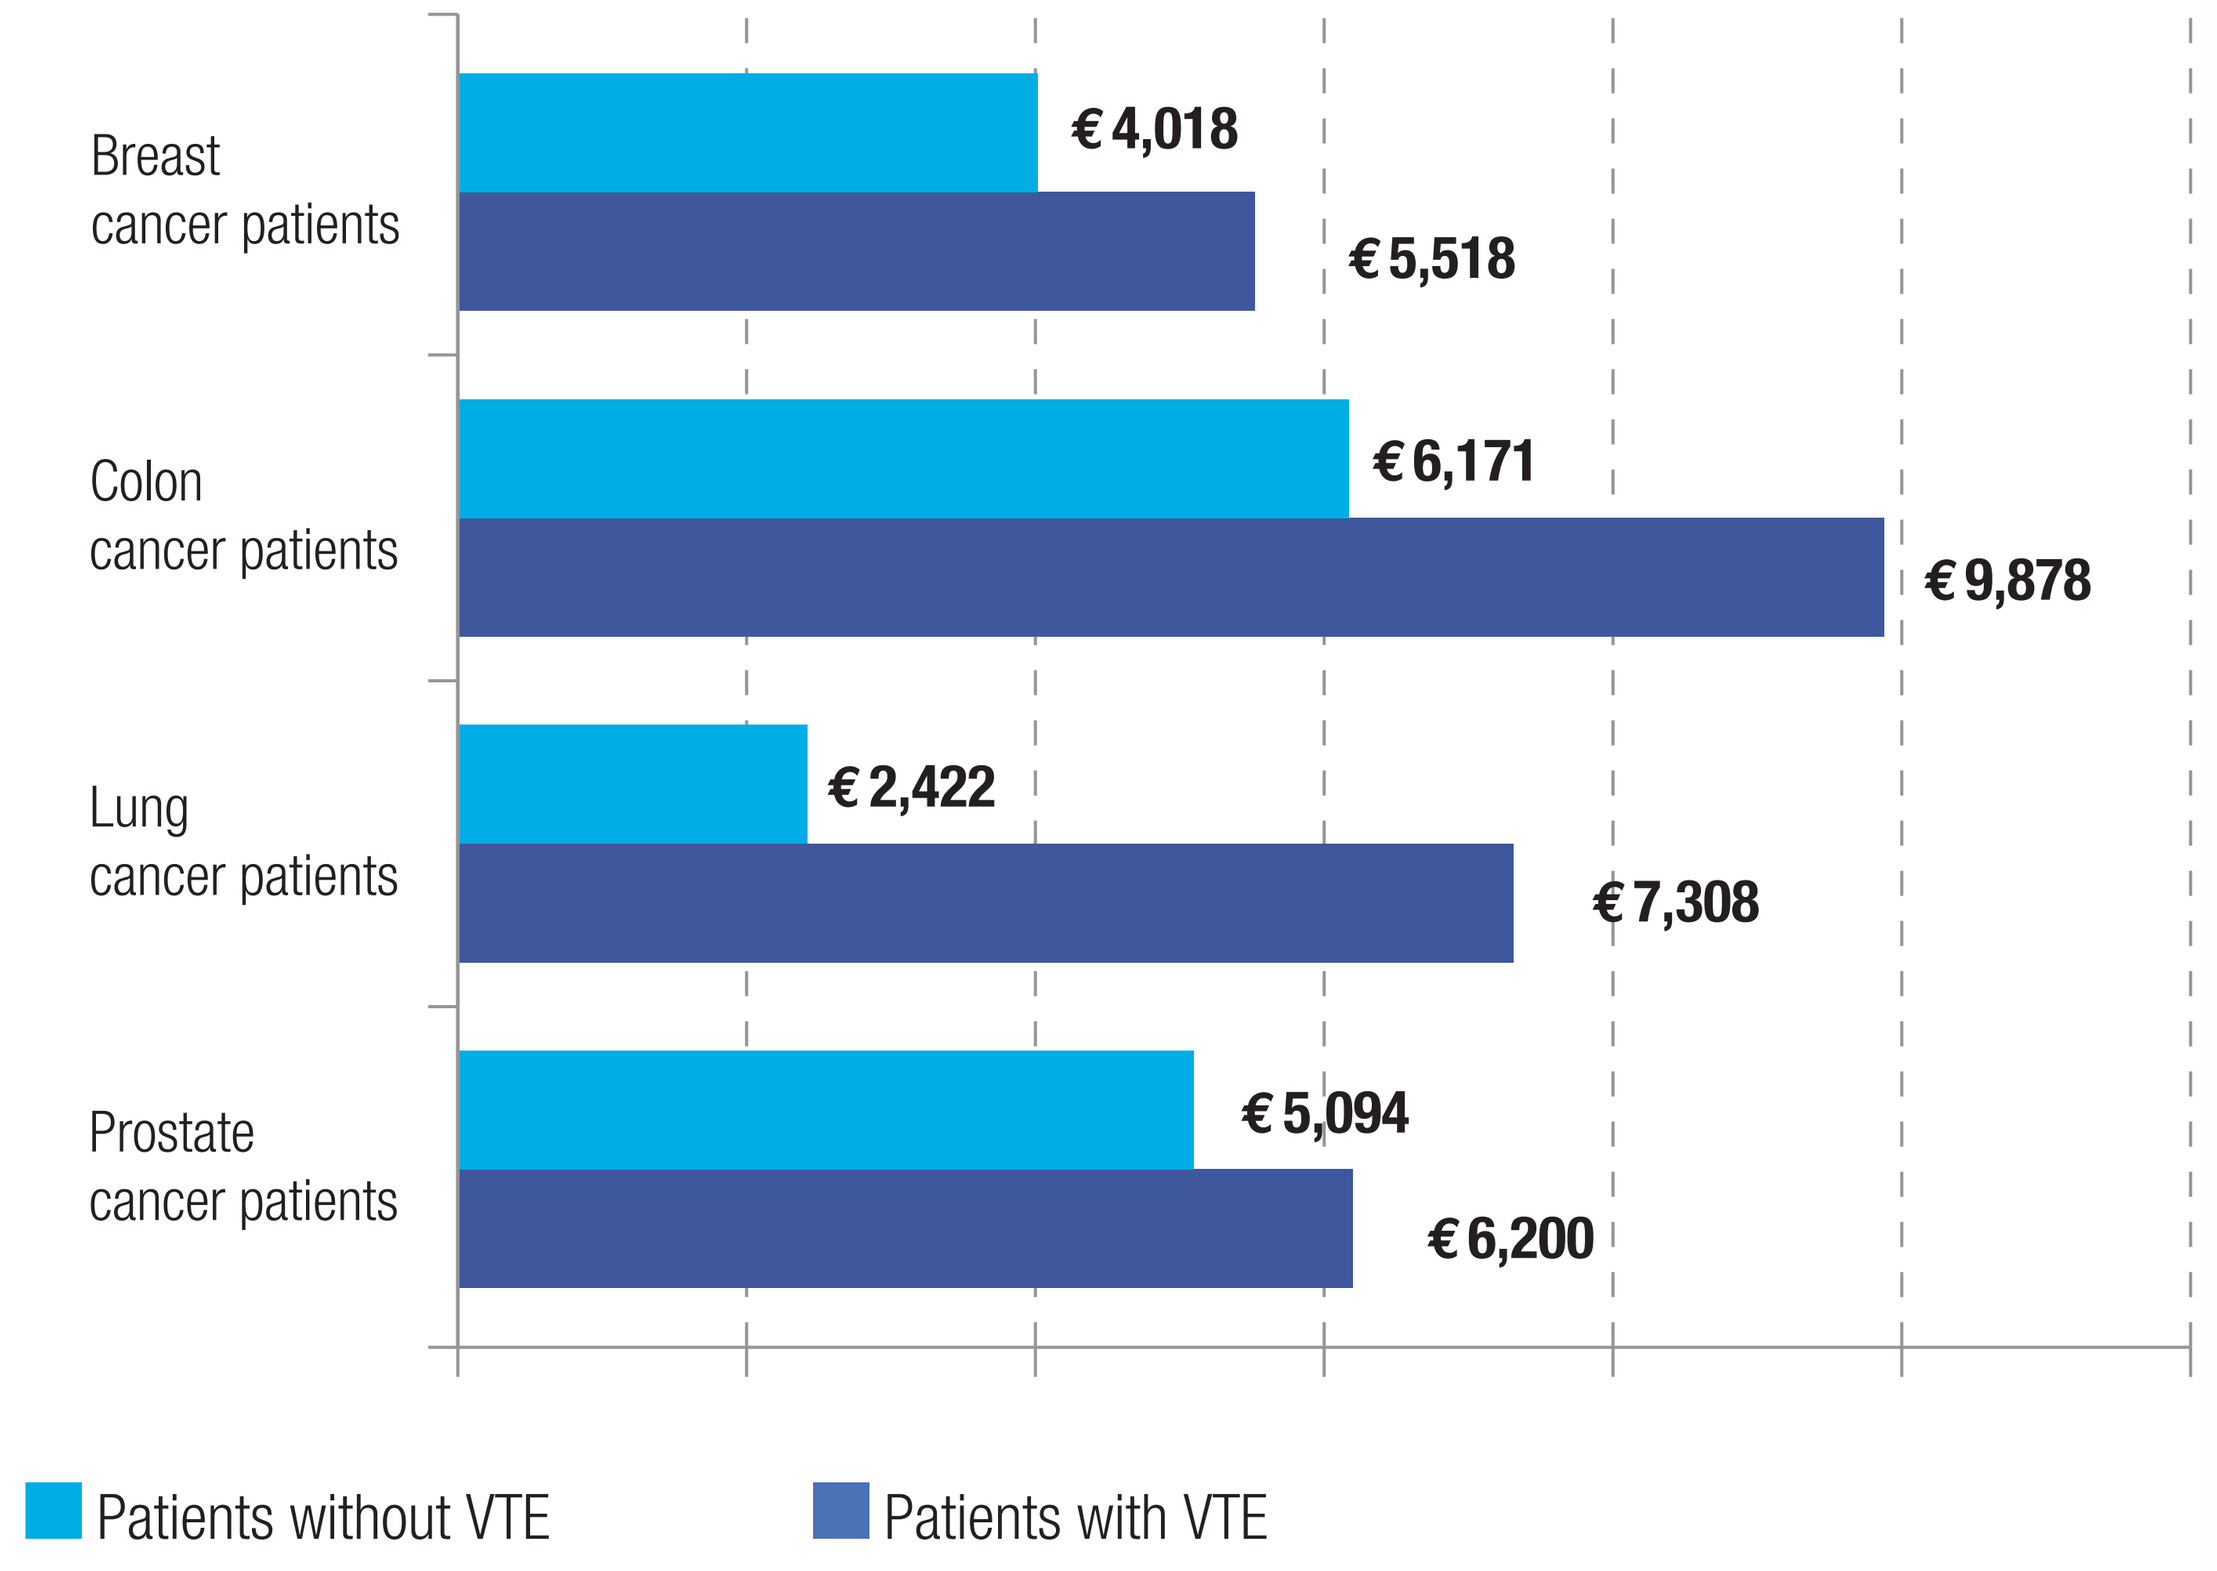
Breast cancer patients
€ 4,018
€ 5,518
Colon cancer patients
€ 6,171
€ 9,878
Lung cancer patients
€ 2,422
€ 7,308
Prostate cancer patients
€ 5,094
€ 6,200
Patients without VTE
Patients with VTE

DISCUSSION / CONCLUSION

- Among a population of 194,964 new patients with breast, colon, lung or prostate cancer, 8,909 patients experienced a thrombosis event corresponding to 12,880 hospitalizations. Around 23.0% of patients who had a first event presented a recurrence, corresponding to 30.8% of total VTE-related admissions. The recurrence rate in our study is in line with previous studies[5,9]. Moreover, recurrent patients experienced the second event in the month following the first occurrence (median time).
- The VTE burden appeared to be high for both patients and healthcare system. In our study, patients admitted for one of the four cancers studied who suffered VTE during hospitalization stayed longer in hospital: median duration of stays almost doubled. Consequently, VTE induced extra hospitalization expenses: the median cost per stay raised from € 4,018 up to € 5,518 in breast cancer, from € 6,171 up to € 9,878 in colon cancer, from € 2,422 up to € 7,308 in lung cancer and from € 5,094 up to € 6,200 in prostate cancer. This cost is substantial considering that the global yearly expenditure for a cancer patient is estimated € 10,000 per year (all types of cancer included)[10].
- Yet, thrombosis incidence could be reduced and recurrence could be avoided by recommended VTE prophylaxis and management. Indeed, although the guidelines are consistent and easy to implement, they are poorly followed:
 - the ENDORSE study showed that only 55% of French cancer patients with thrombosis risks were administered prophylaxis (2011)[11];
 - the CARMEN study showed that 58% of French cancer patients with VTE were treated in accordance with recommendations (2010)[12].
- Thus, a good follow-up of recommendations, adequate and prolonged curative treatment as well as ambulatory management could reduce VTE-related costs as well as stay's duration and benefit both patients and healthcare payer.

1. EOCD. Panorama de la santé 2011 : les indicateurs de l'OCDE. 2011.
2. Chew HK *et al.* Incidence of venous thromboembolism and its effect on survival among patients with common cancers. Archives of Internal Medicine 2006.
3. MacLellan DG *et al.* Venous thromboembolism and cancer. ANZ Journal of Surgery 2012.
4. Lyman GH *et al.* Venous thromboembolism risk in patients with cancer receiving chemotherapy: a real-world analysis. Oncologist 2013.
5. Prandoni P. *et al.* Recurrent venous thromboembolism and bleeding complications during anticoagulant treatment in patients with cancer and venous thrombosis. Blood 2002.
6. Khorana AA. *et al.* Health care costs associated with venous thromboembolism in selected high-risk ambulatory patients with solid tumors undergoing chemotherapy in the United States. Clinicoecon Outcomes Res 2013.
7. Scotté F *et al.* The cost of thrombolic events in hospitalized patients with breast or prostate cancer in France. Advances in Therapy. 2015
8. Hassett MJ *et al.* Frequency and cost of chemotherapy-related serious adverse effects in a population sample of women with breast cancer. Journal of the National Cancer Institute 2006.
9. Yu YB *et al.* A nation-wide analysis of venous thromboembolism in 497,180 cancer patients with the development and validation of a risk-stratification scoring system. Thrombosis and Haemostasis 2012.
10. Bullano *et al.* Longitudinal evaluation of health plan cost per venous thromboembolism or bleed event in patients with a prior venous thromboembolism event during hospitalization. J Manag Care Pharm 2005.
11. Bergmann JL-L. *et al.* Venous thromboembolism risk and prophylaxis in the acute hospital care setting (ENDORSE study): results obtained in France. Presse Medicale 2011.
12. Sevestre MB. *et al.* Compliance of recommendations with French clinical practice in the management of thromboembolism in patients with cancer: The CARMEN study. J Clin Oncol 2012.

Contact with Dr F. Scotté : florian.scotte@aphp.fr

Sponsored by LEO Pharma

RESERVE QR CODE